Bioprinted three-dimensional (3D) human liver constructs provide a model for interrogating liver biology

Justin B. Robbins, Colin M. O'Neill, Vivian Gorgen, Sharon C. Presnell, and Benjamin R. Shepherd
Tissue Applications and Systems Engineering Groups
Organovo, Inc., 6275 Nancy Ridge Dr., Suite 110, San Diego, CA, USA

Presentation Number: 2176

Abstract

Conventional two dimensional (2D) cell culture models do not reflect the complexity of biological phenomena that occur in the liver microenvironment, resulting in discrepancies between in vitro predictions and in vivo realities. One of the major reasons for these differences is that primary hepatocytes, when isolated and cultured in a 2D monolayer, rapidly lose key enzymatic properties that are considered essential for predictive modeling. Loss of these key phenotypic features is one of the major limitations in toxicology screening protocols involving human hepatocytes, which has slowed the pace of development for new drug entities. In addition to challenges in maintaining differentiation, species-specific differences in hepatocellular function have also been extensively reported, making it difficult to extrapolate non-human data to the clinic. Therefore, providing better in vitro models for interrogating human liver biology will have a major impact in the fields of toxicology and hepatology. In this report, we demonstrate that 3D bioprinted liver tissue mimetics composed of parenchymal (human primary hepatocytes and hepatic cell lines) and non-parenchymal cell populations (endothelial cells [EC] and hepatic stellates) can be cultured for up to 42 days and retain key liver features and functions. The 3D liver neotissues are metabolically active for at least 42 days, producing essential liver-derived serum proteins including albumin, fibrinogen, and transferrin within the medium. Histological analysis of formalin-fixed paraffin-embedded tissues at multiple time points revealed well-organized architecture, with intercellular junctions between parenchymal cells and clear evidence of lumenized, CD31-positive, EC-lined microvascular structures. Toxicity studies by examining ATP and lactate dehydrogenase (LDH) activities of the 3D liver constructs when dosed with known hepatotoxic agents were undertaken. Exposure of the bioprinted liver tissue to acetaminophen resulted in LD_{50} values similar to published human in vivo values. Enhancement of the acetaminophen toxic effect was observed when combined with ethanol treatment of bioprinted neotissues. Finally, the 3D liver neotissues responded in a dose dependent manner to the small molecule diclofenac, a known hepatotoxin. Not only do 3D neotissues allow biochemical interrogation of these toxicants, they also permit physical examination of the cellular populations within the tissue by histology. These results demonstrate the potential for the bioprinted 3D liver tissues in drug discovery and development, and human disease modeling.

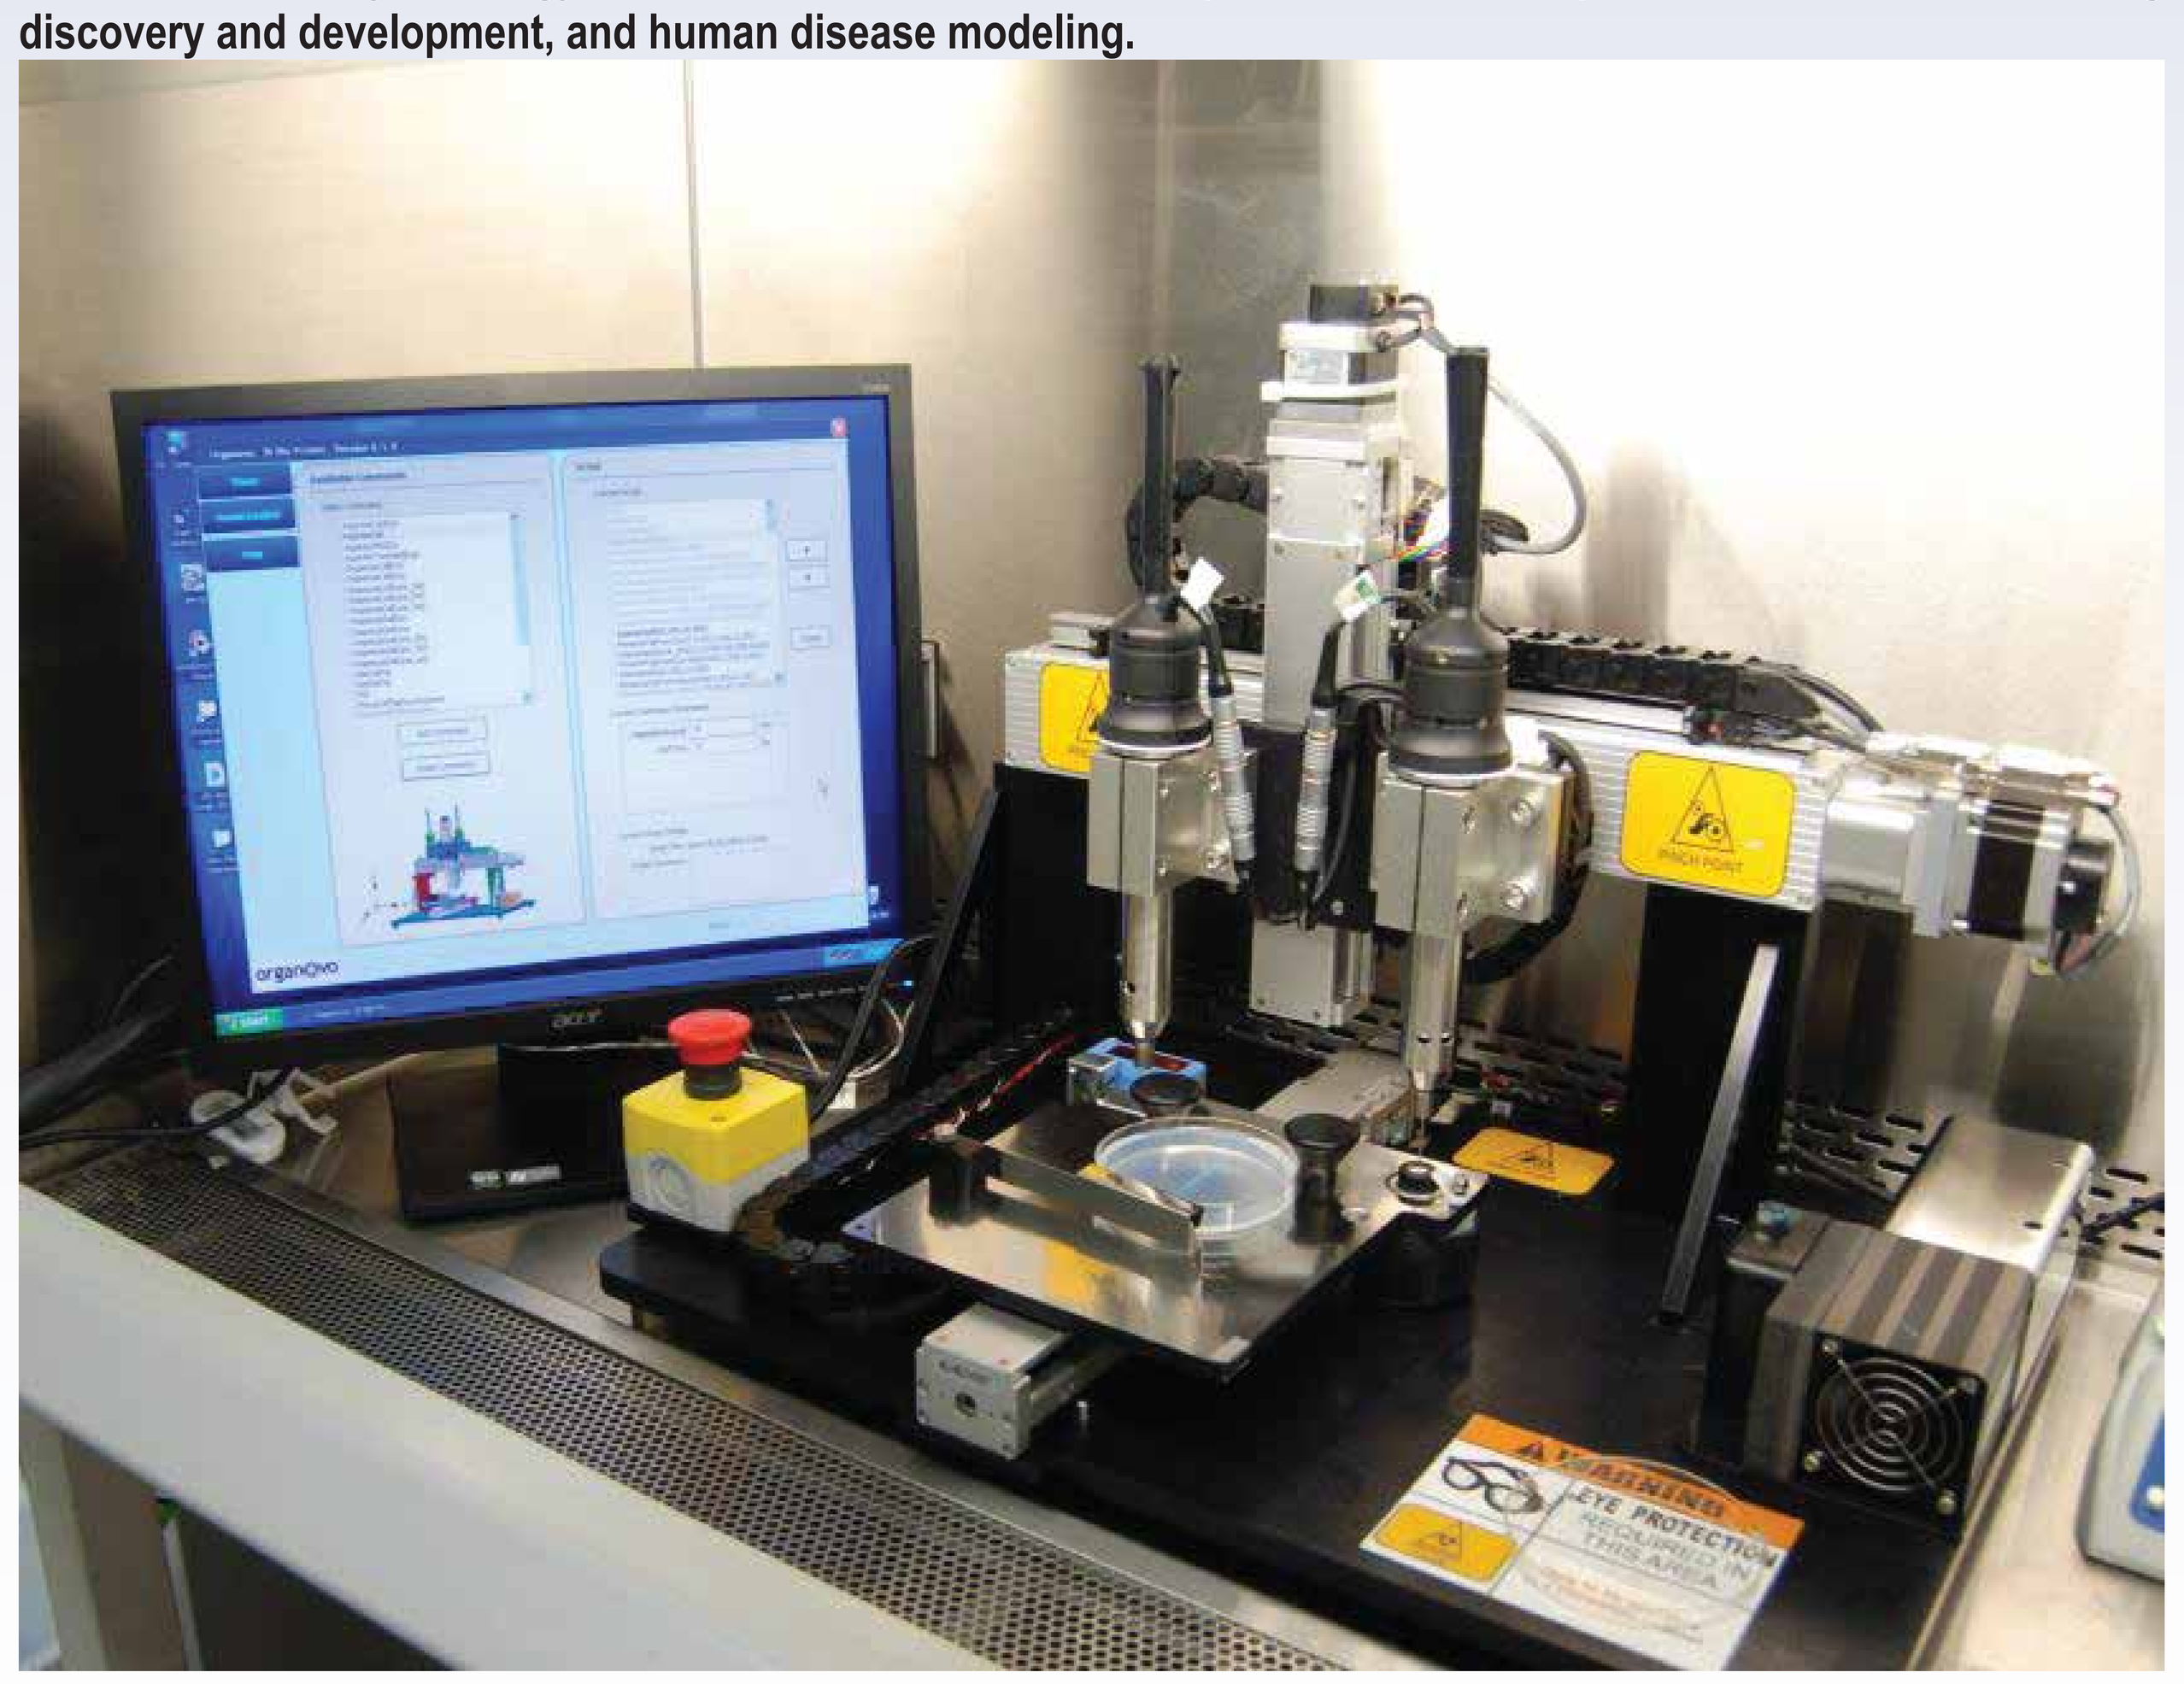

Figure 1. Novogen MMX Bioprinter™

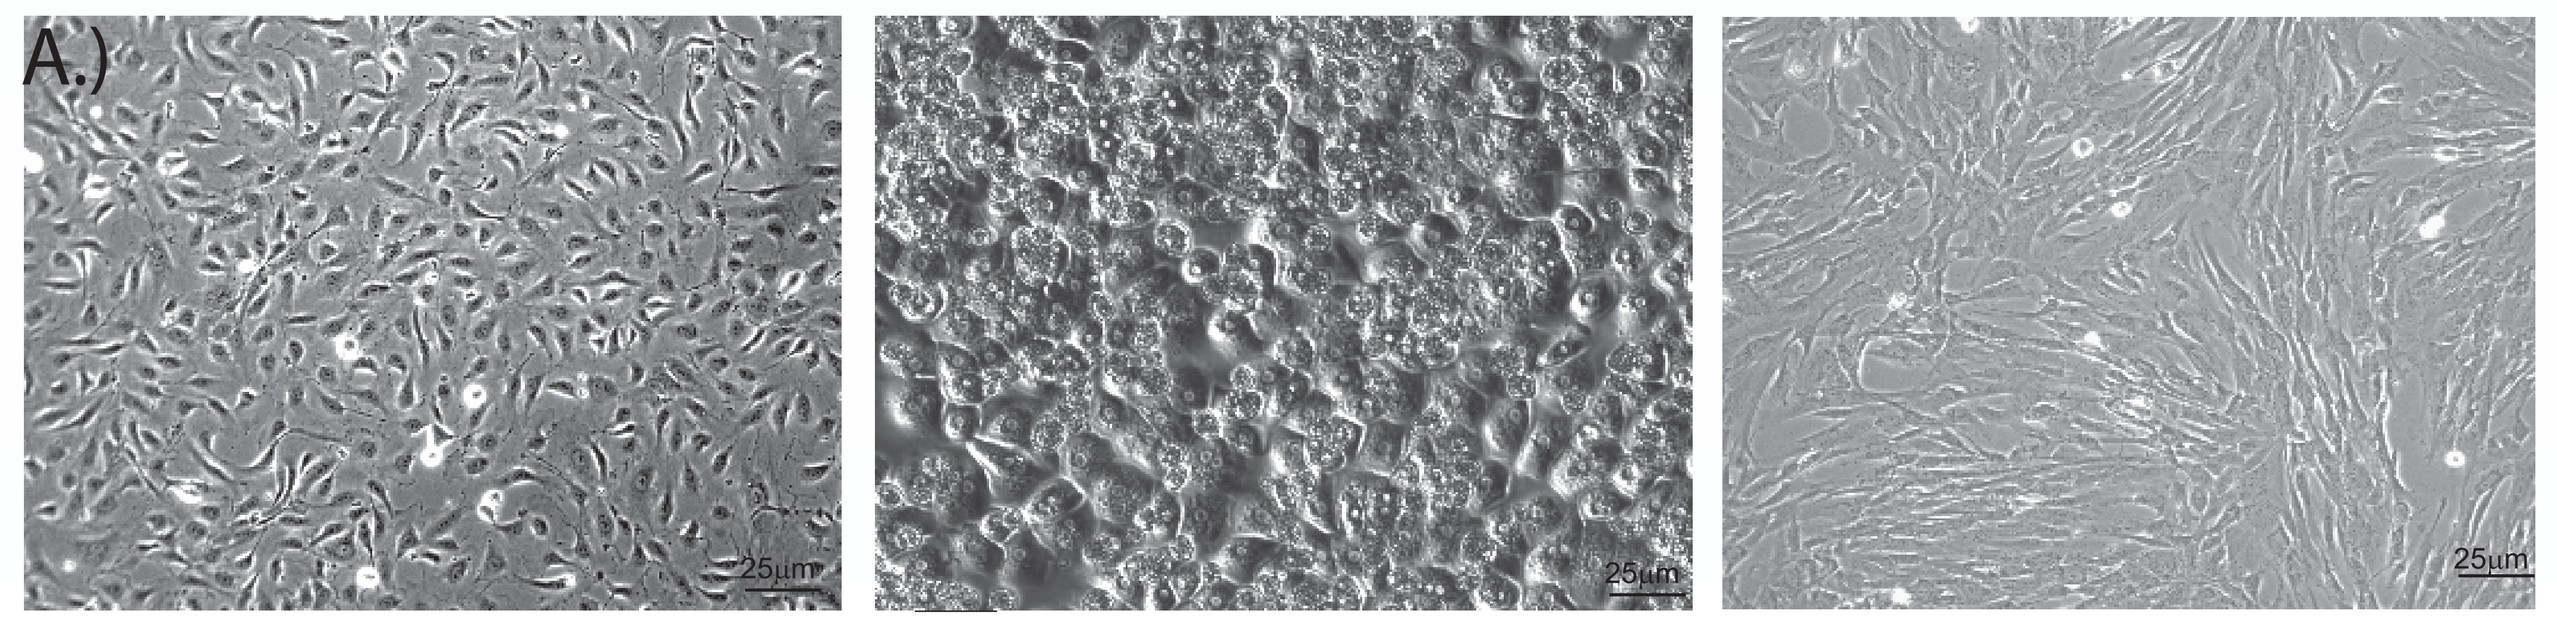
A.)
25µm
25µm
25µm

Human Endothelial Cells **Human Hepatocytes** **Human Hepatic Stellates**

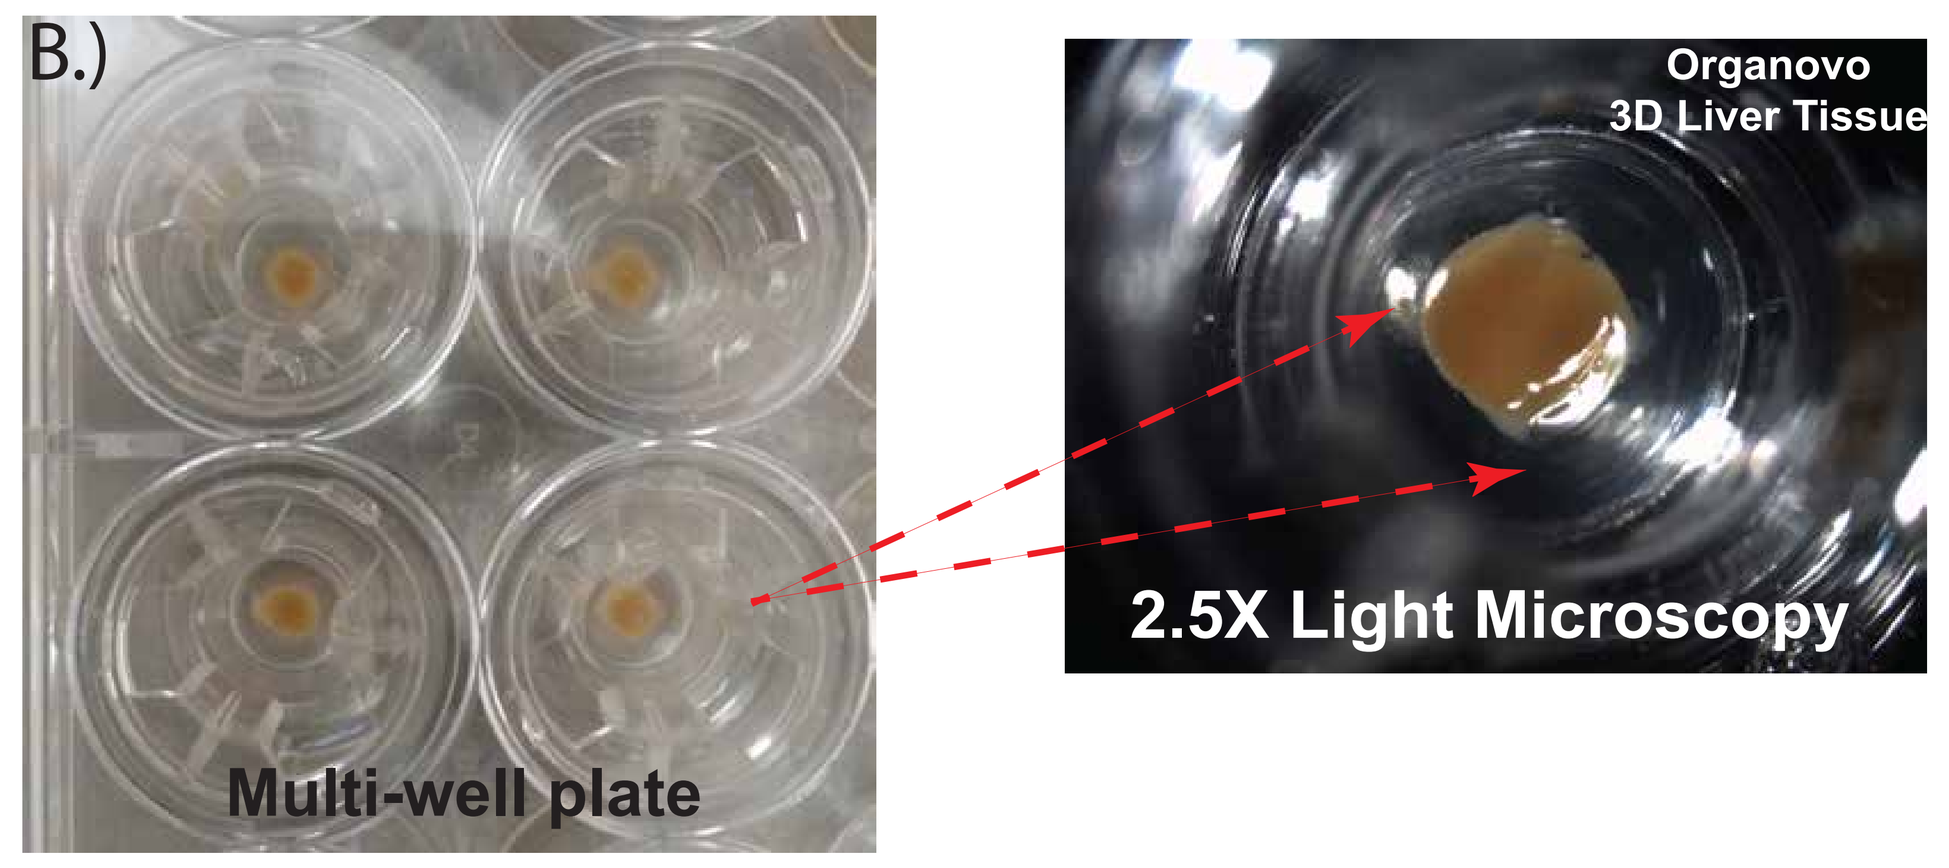
B.)
Organovo
3D Liver Tissue
2.5X Light Microscopy
Multi-well plate

Figure 2. Features of Organovo's 3D bioprinted human liver tissues. A.) Bioprinted 3D human liver tissues include parenchymal and non-parenchymal cell types deposited in distinct compartments. B.) Multi-well format enables compound screening.

Results

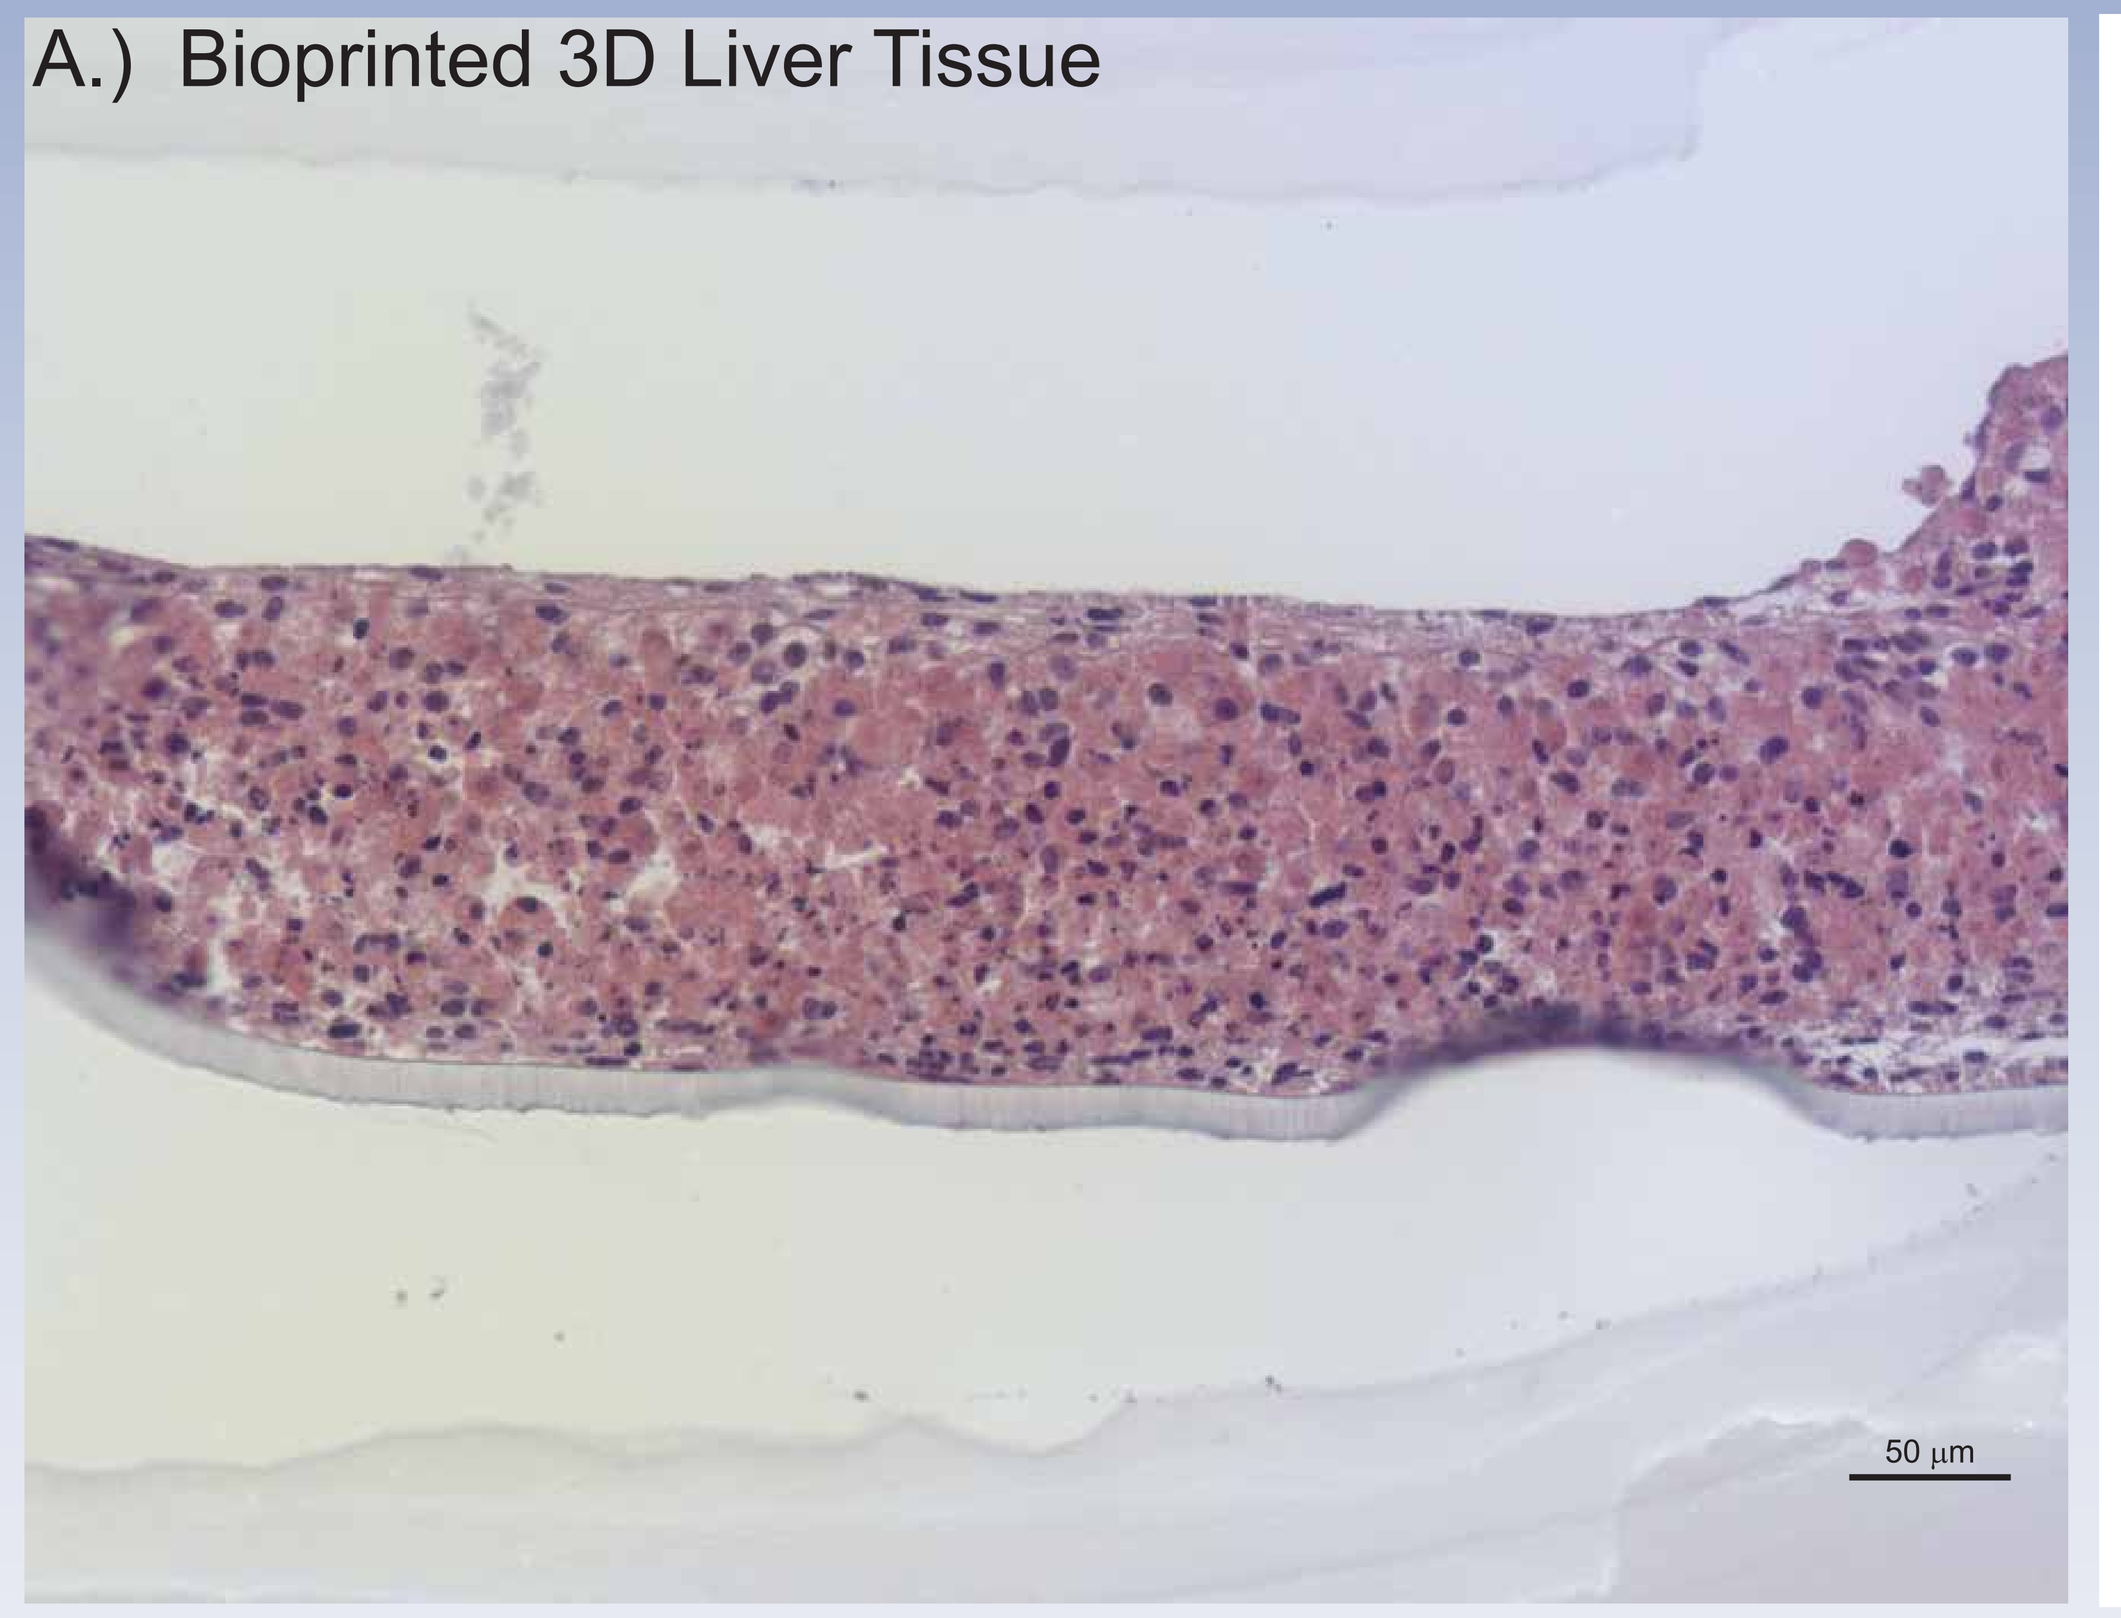
A.) Bioprinted 3D Liver Tissue
50 µm

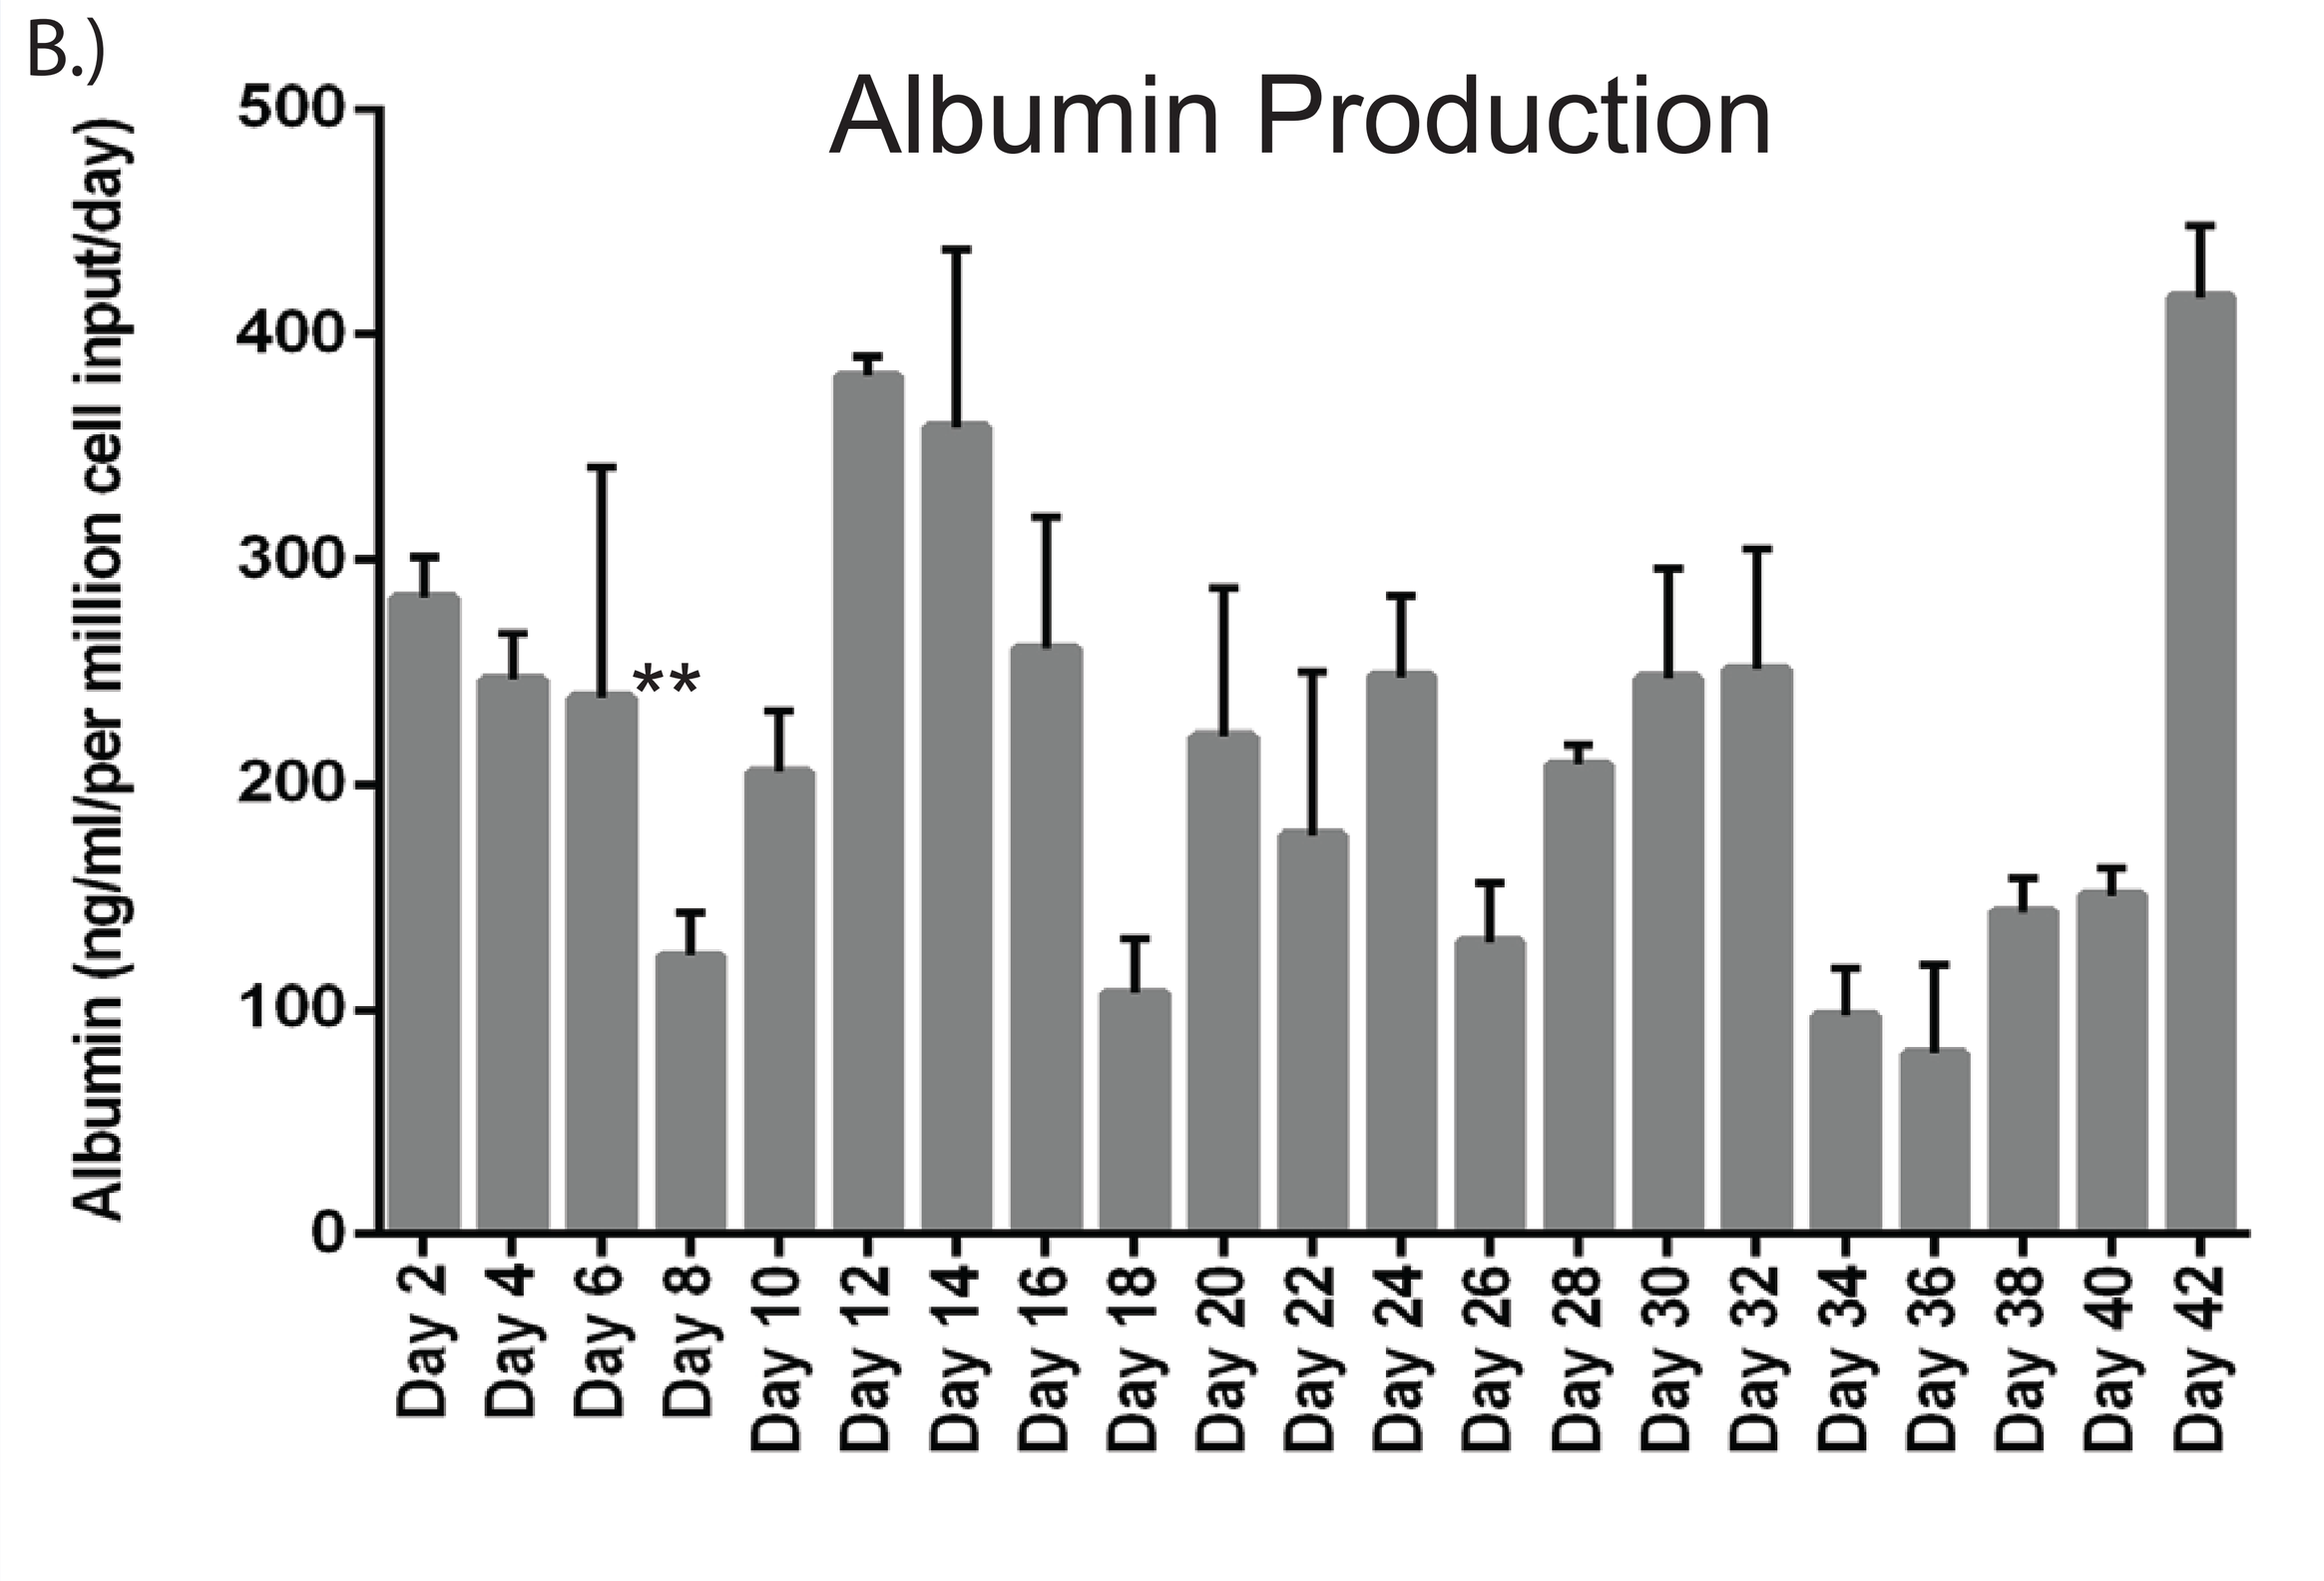
B.)
Albumin Production
Albumin (ng/ml/per million cell input/day)
0
100
200
300
400
500
**
Day 2
Day 4
Day 6
Day 8
Day 10
Day 12
Day 14
Day 16
Day 18
Day 20
Day 22
Day 24
Day 26
Day 28
Day 30
Day 32
Day 34
Day 36
Day 38
Day 40
Day 42

Figure 3. 3D bioprinted tissues show tissue-like perfomance over extended time periods. A.) Bioprinted tissues exhibit dense tissue-like architecture demonstrated by H&E staining at day 7 (**). B.) Performance of 3D liver tissues over 42 days was validated by measuring albumin production over time.

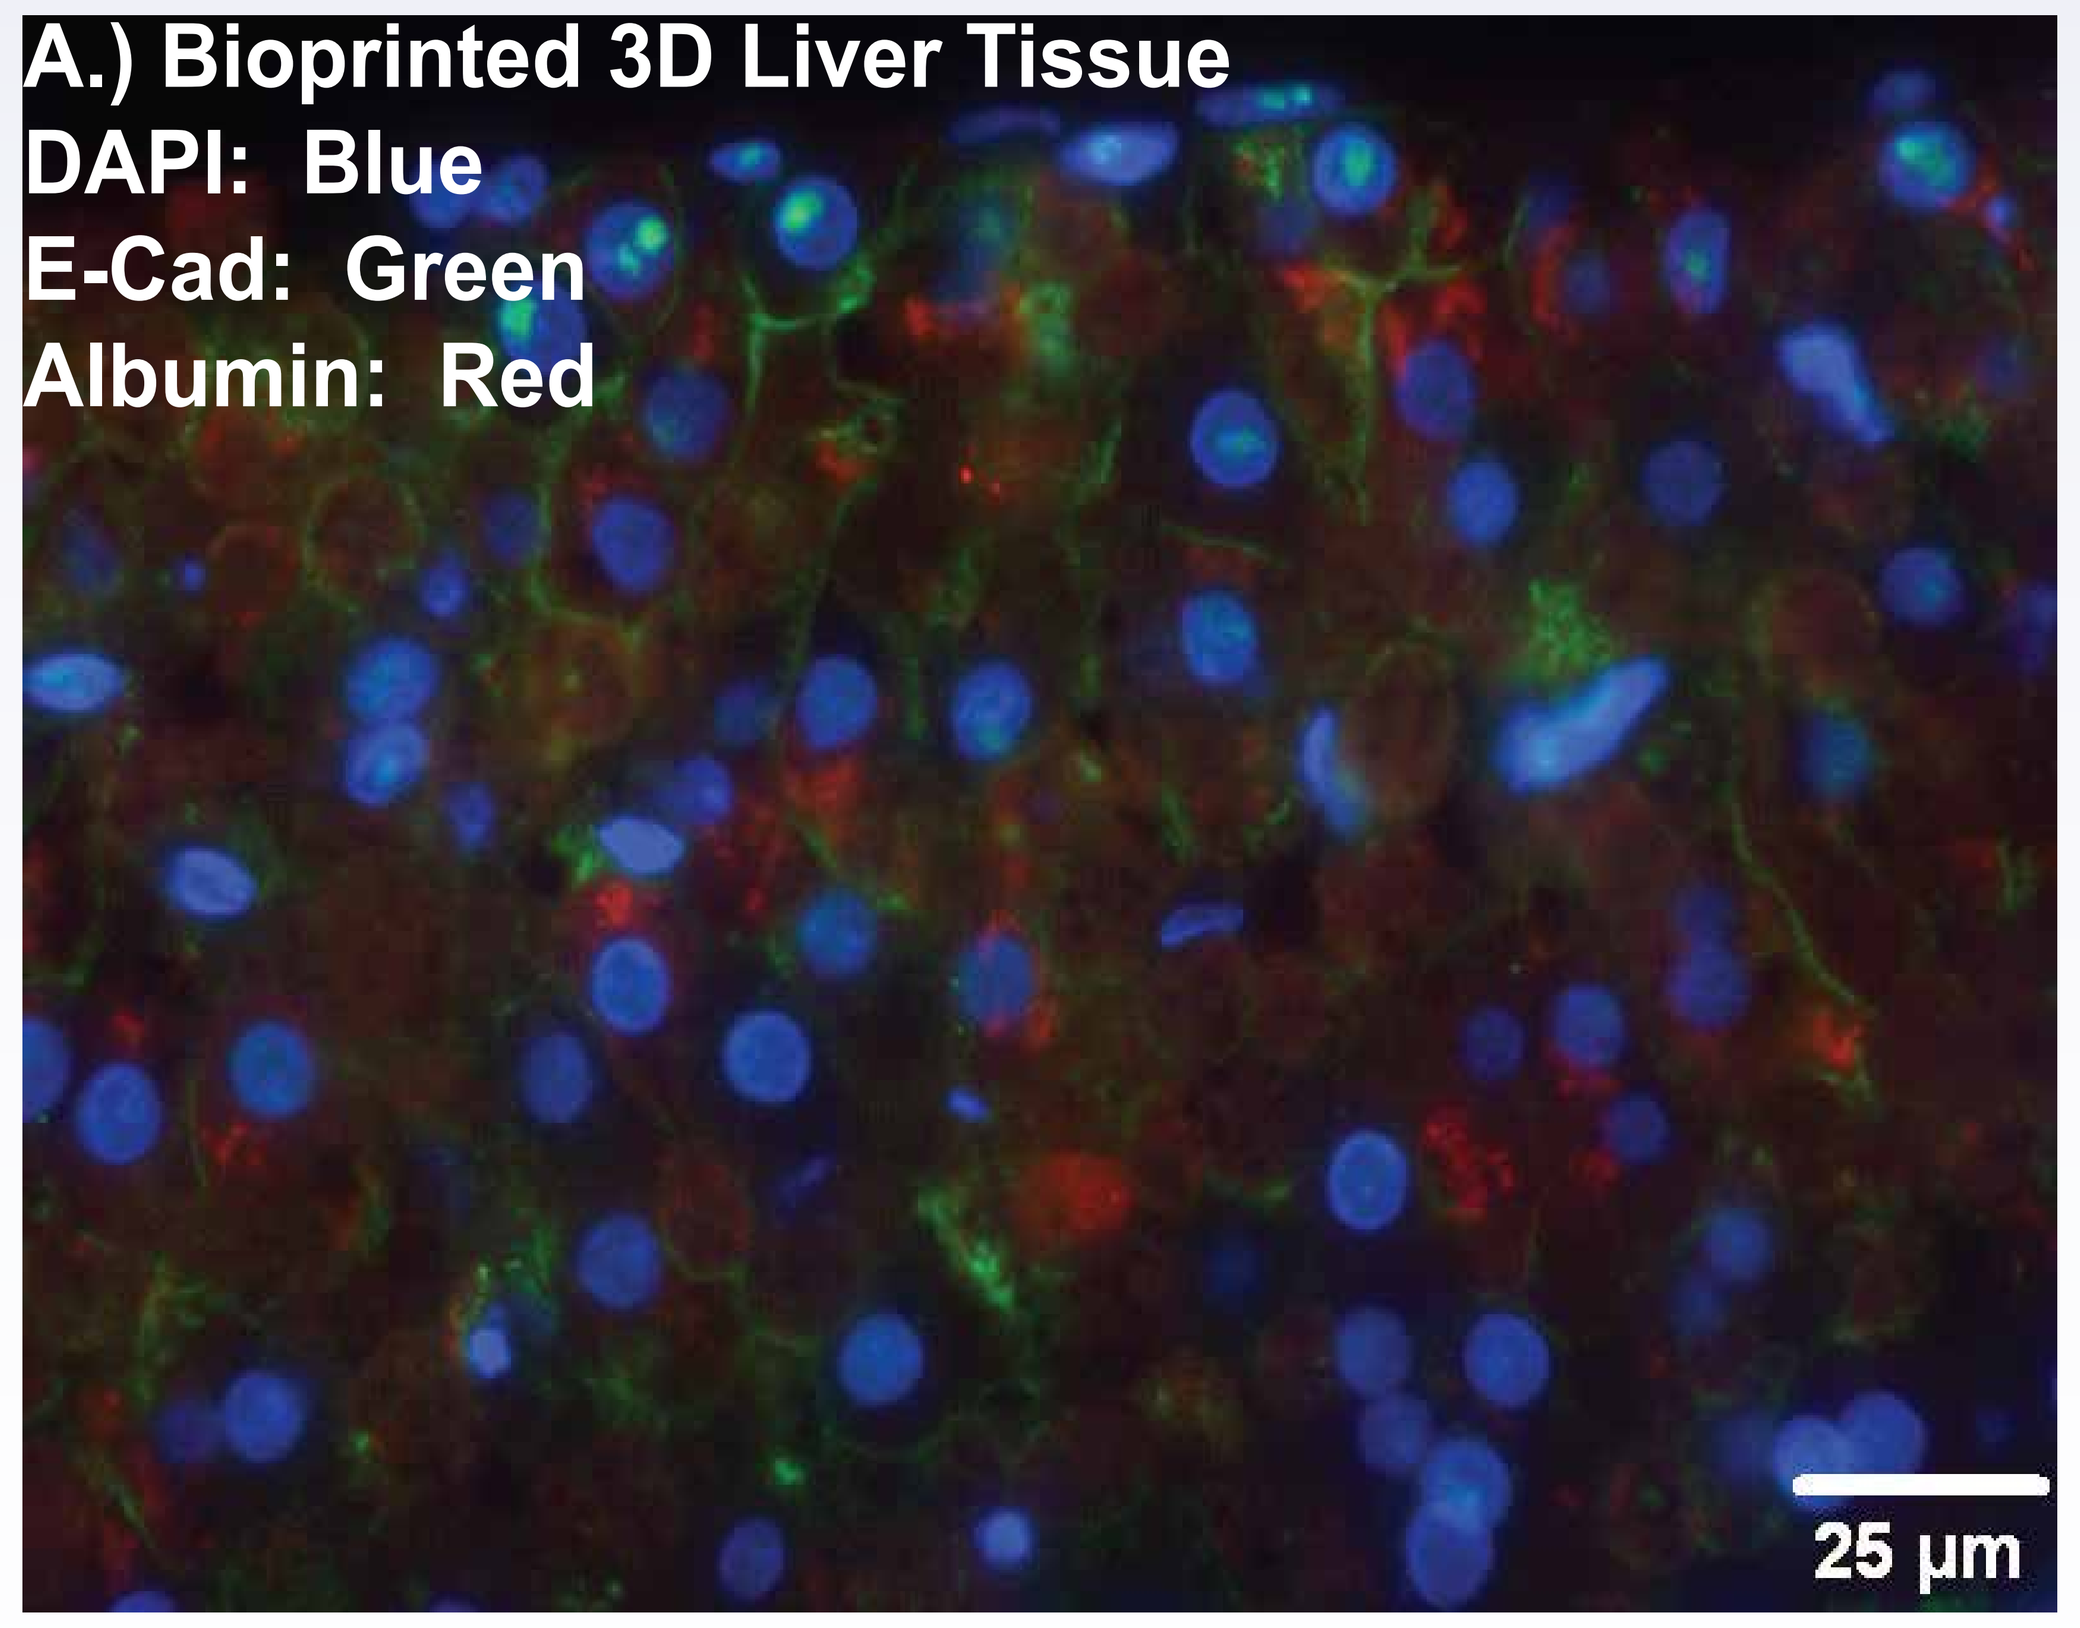
A.) Bioprinted 3D Liver Tissue
DAPI: Blue
E-Cad: Green
Albumin: Red
25 µm

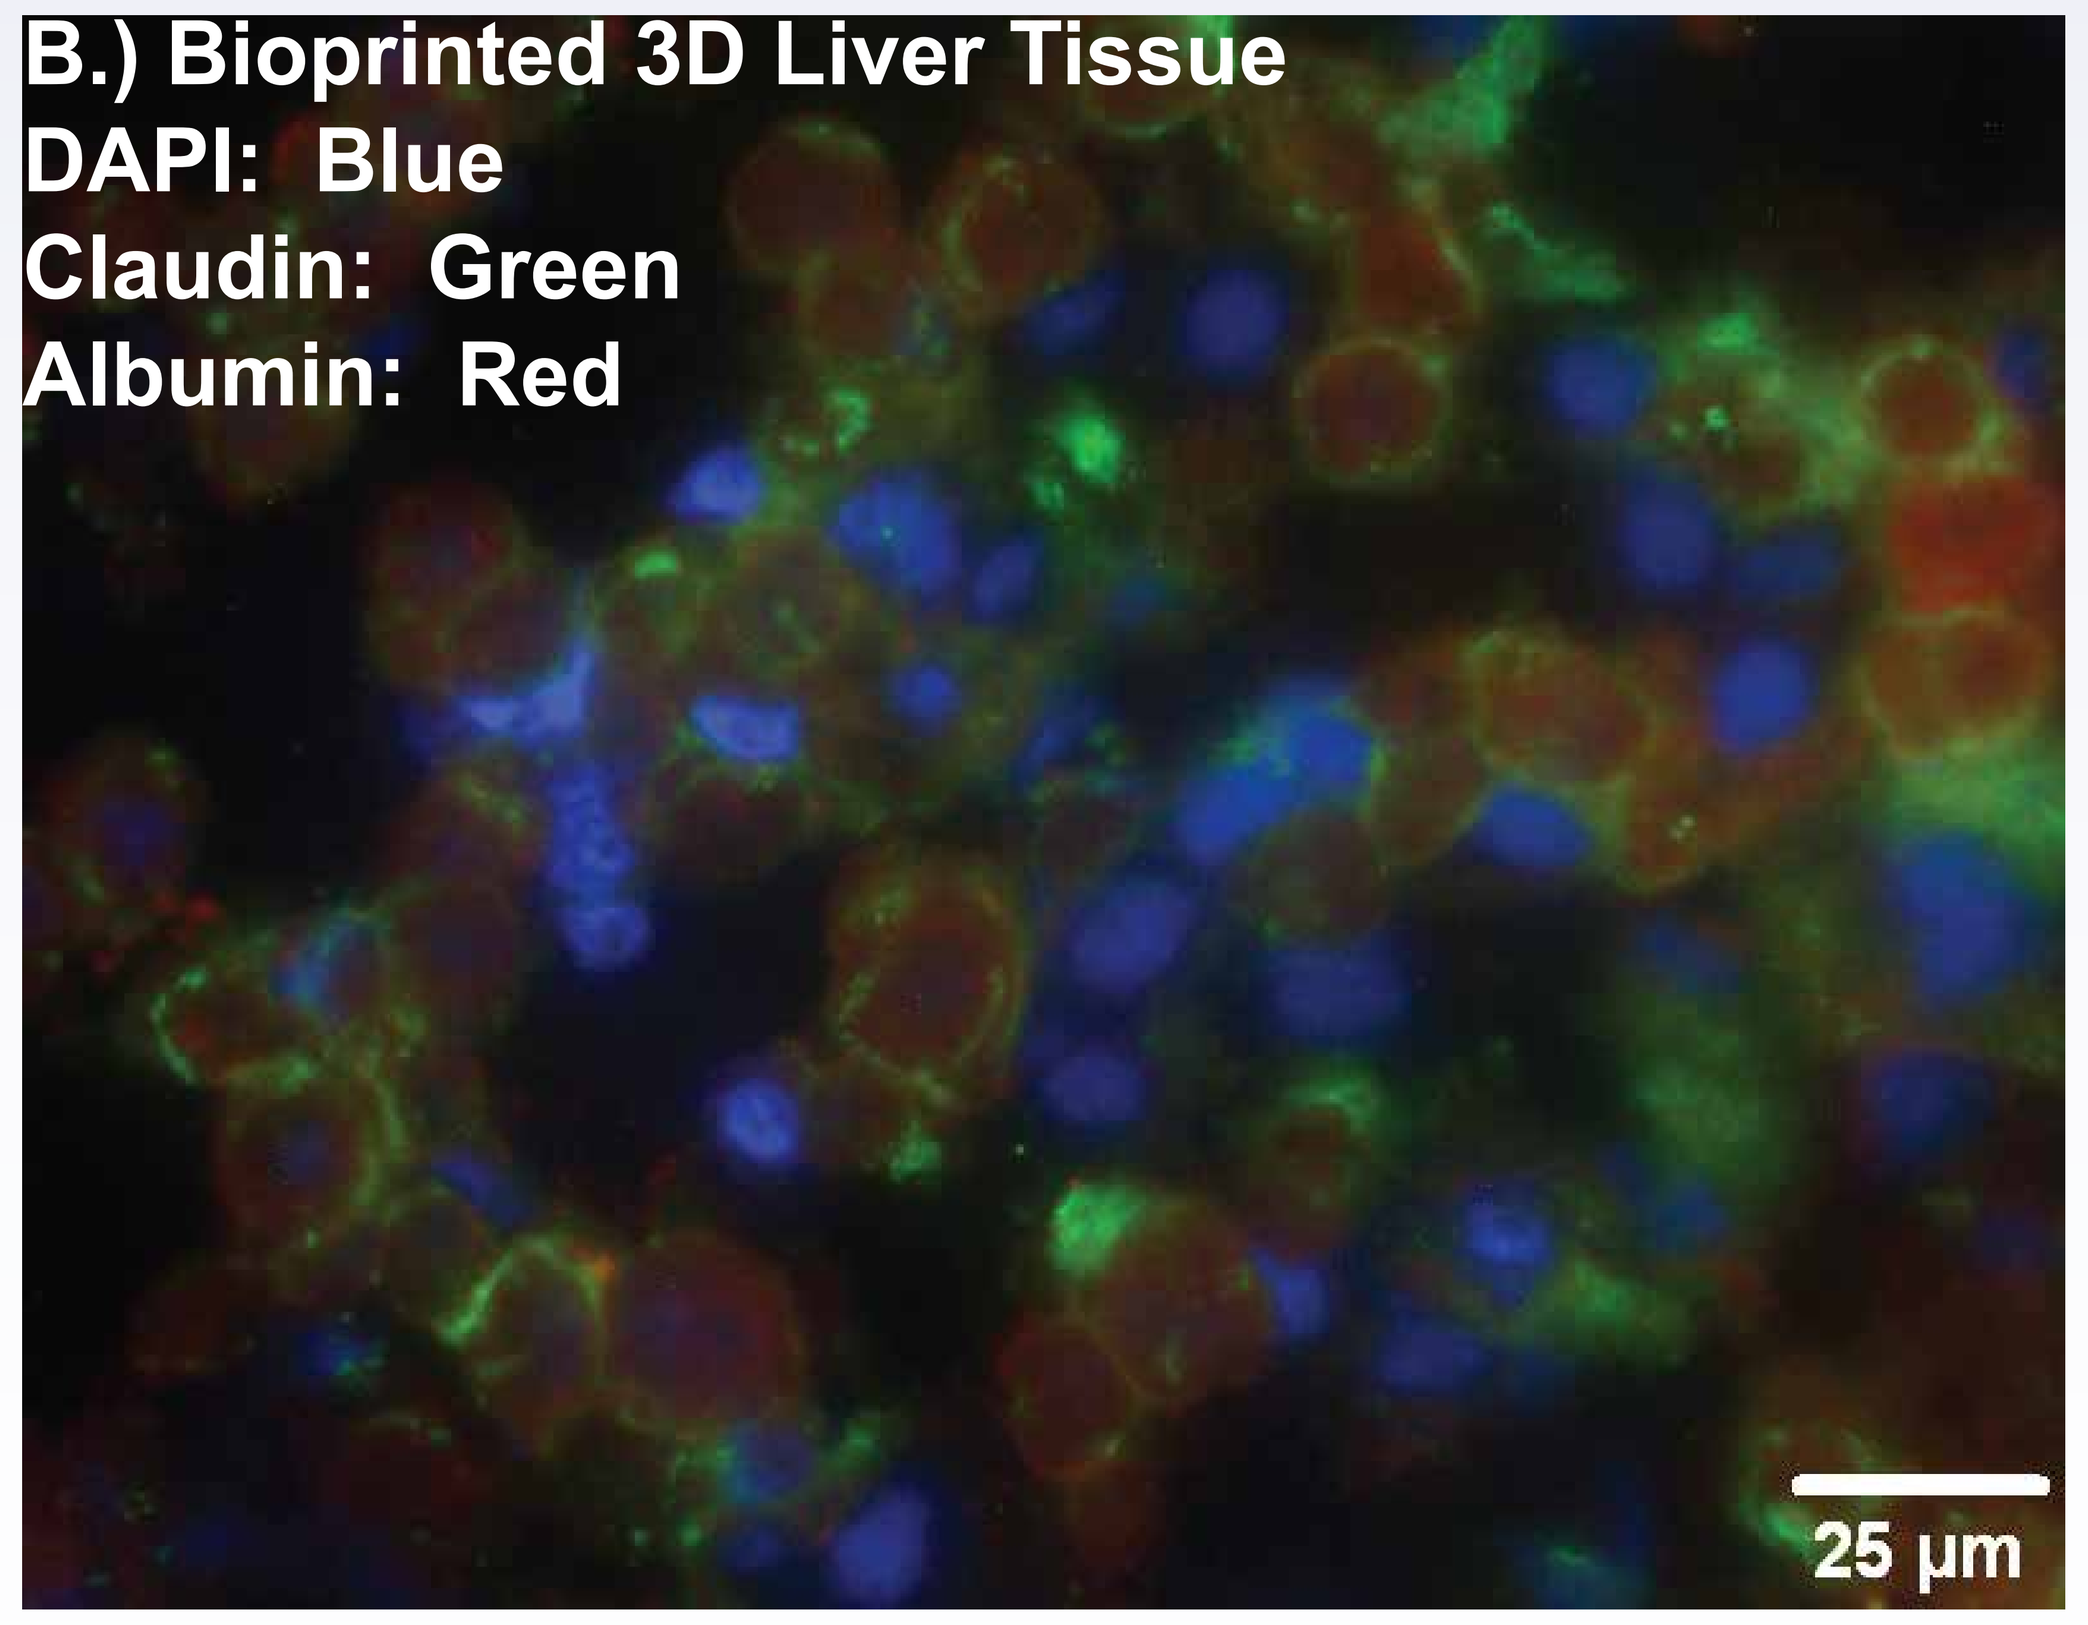
B.) Bioprinted 3D Liver Tissue
DAPI: Blue
Claudin: Green
Albumin: Red
25 µm

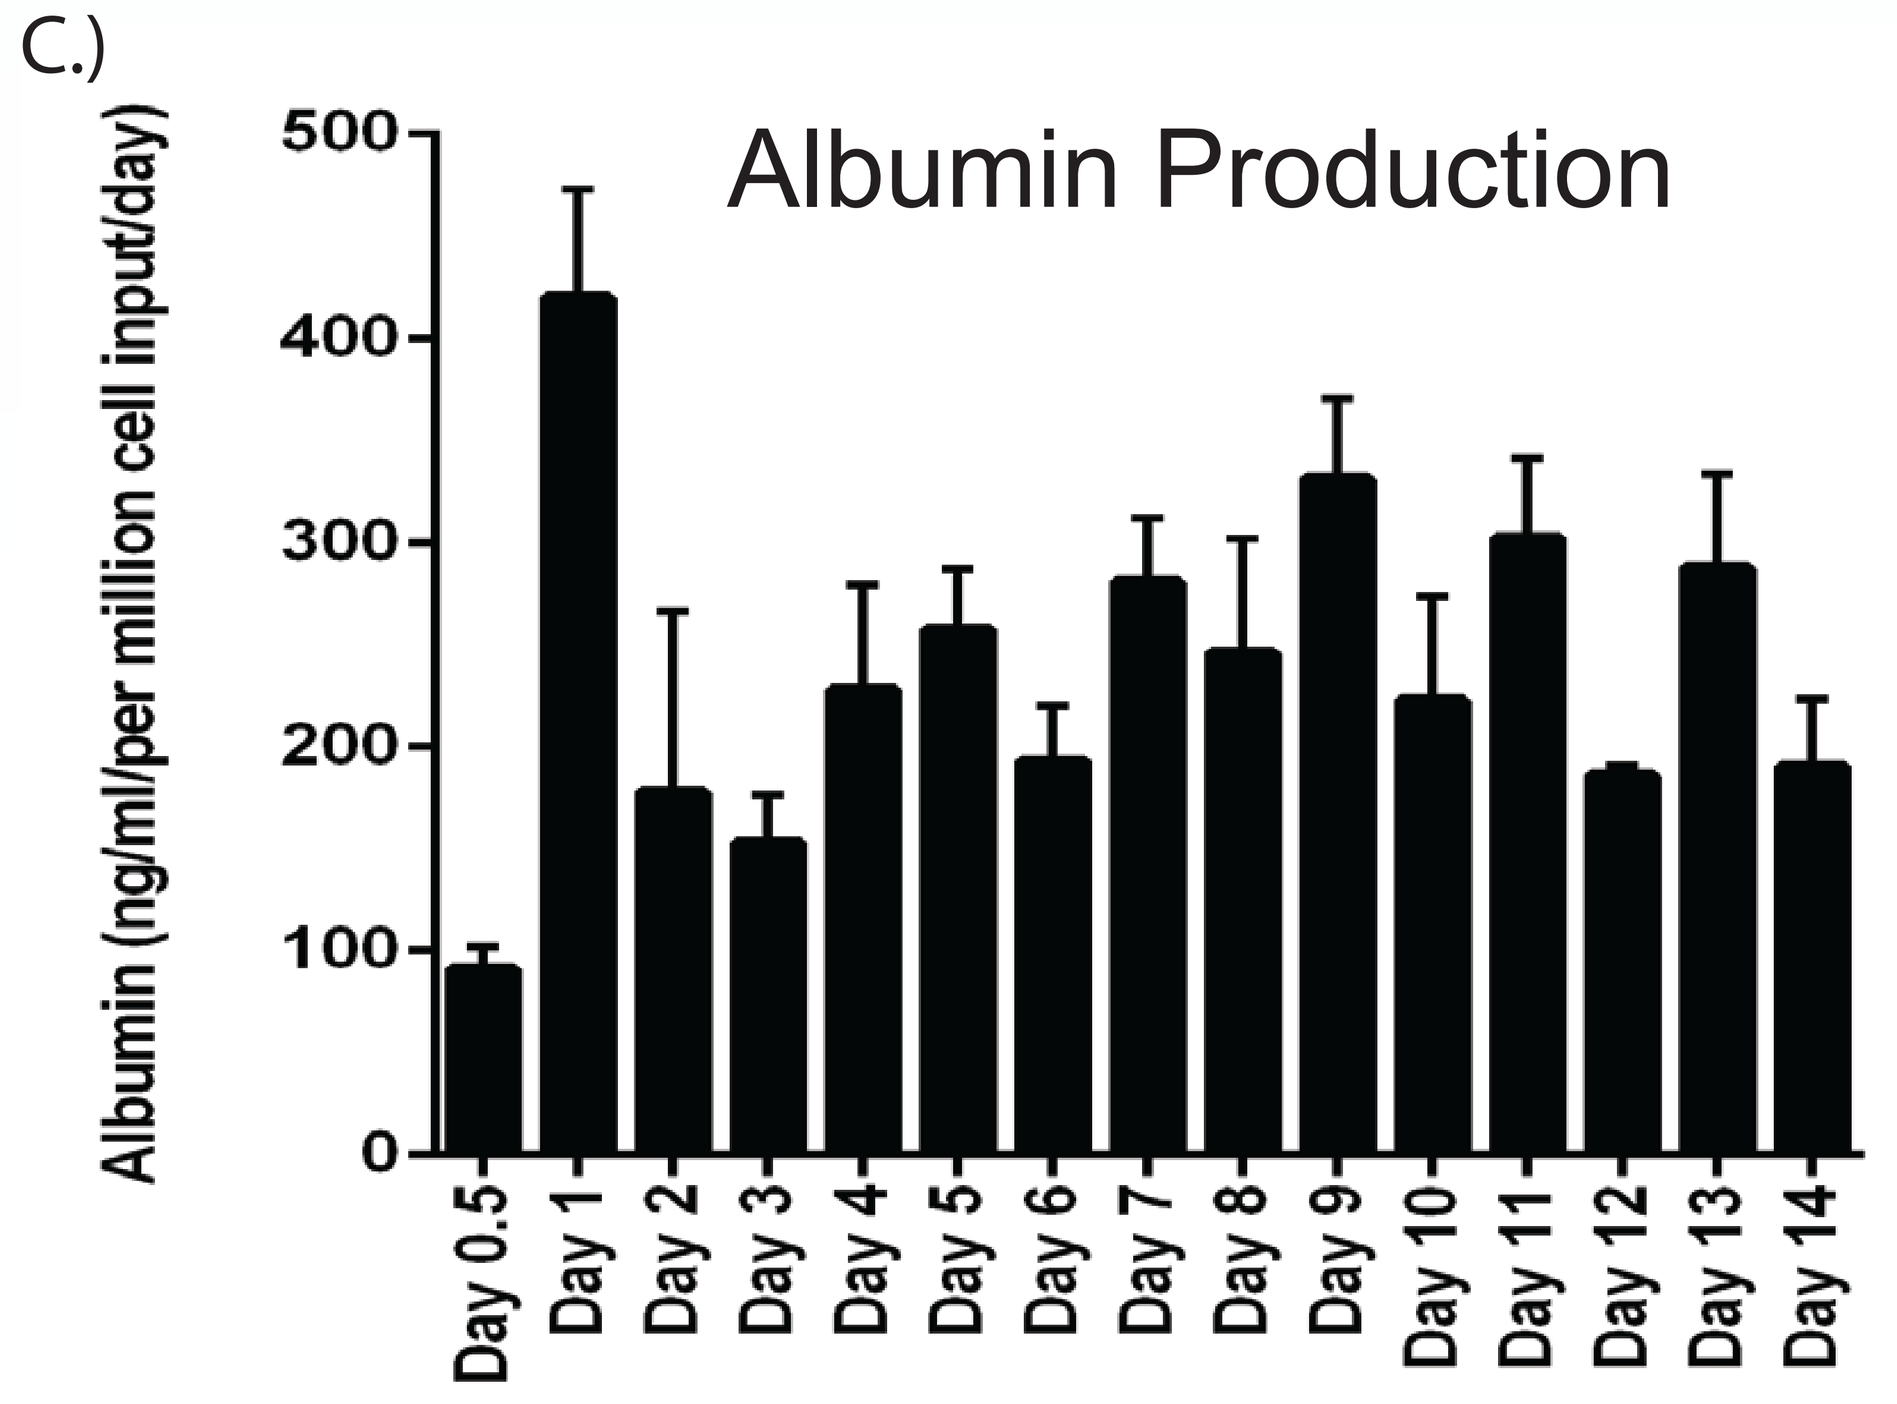
C.)
Albumin Production
Albumin (ng/ml/per million cell input/day)
0
100
200
300
400
500
Day 0.5
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Day 7
Day 8
Day 9
Day 10
Day 11
Day 12
Day 13
Day 14

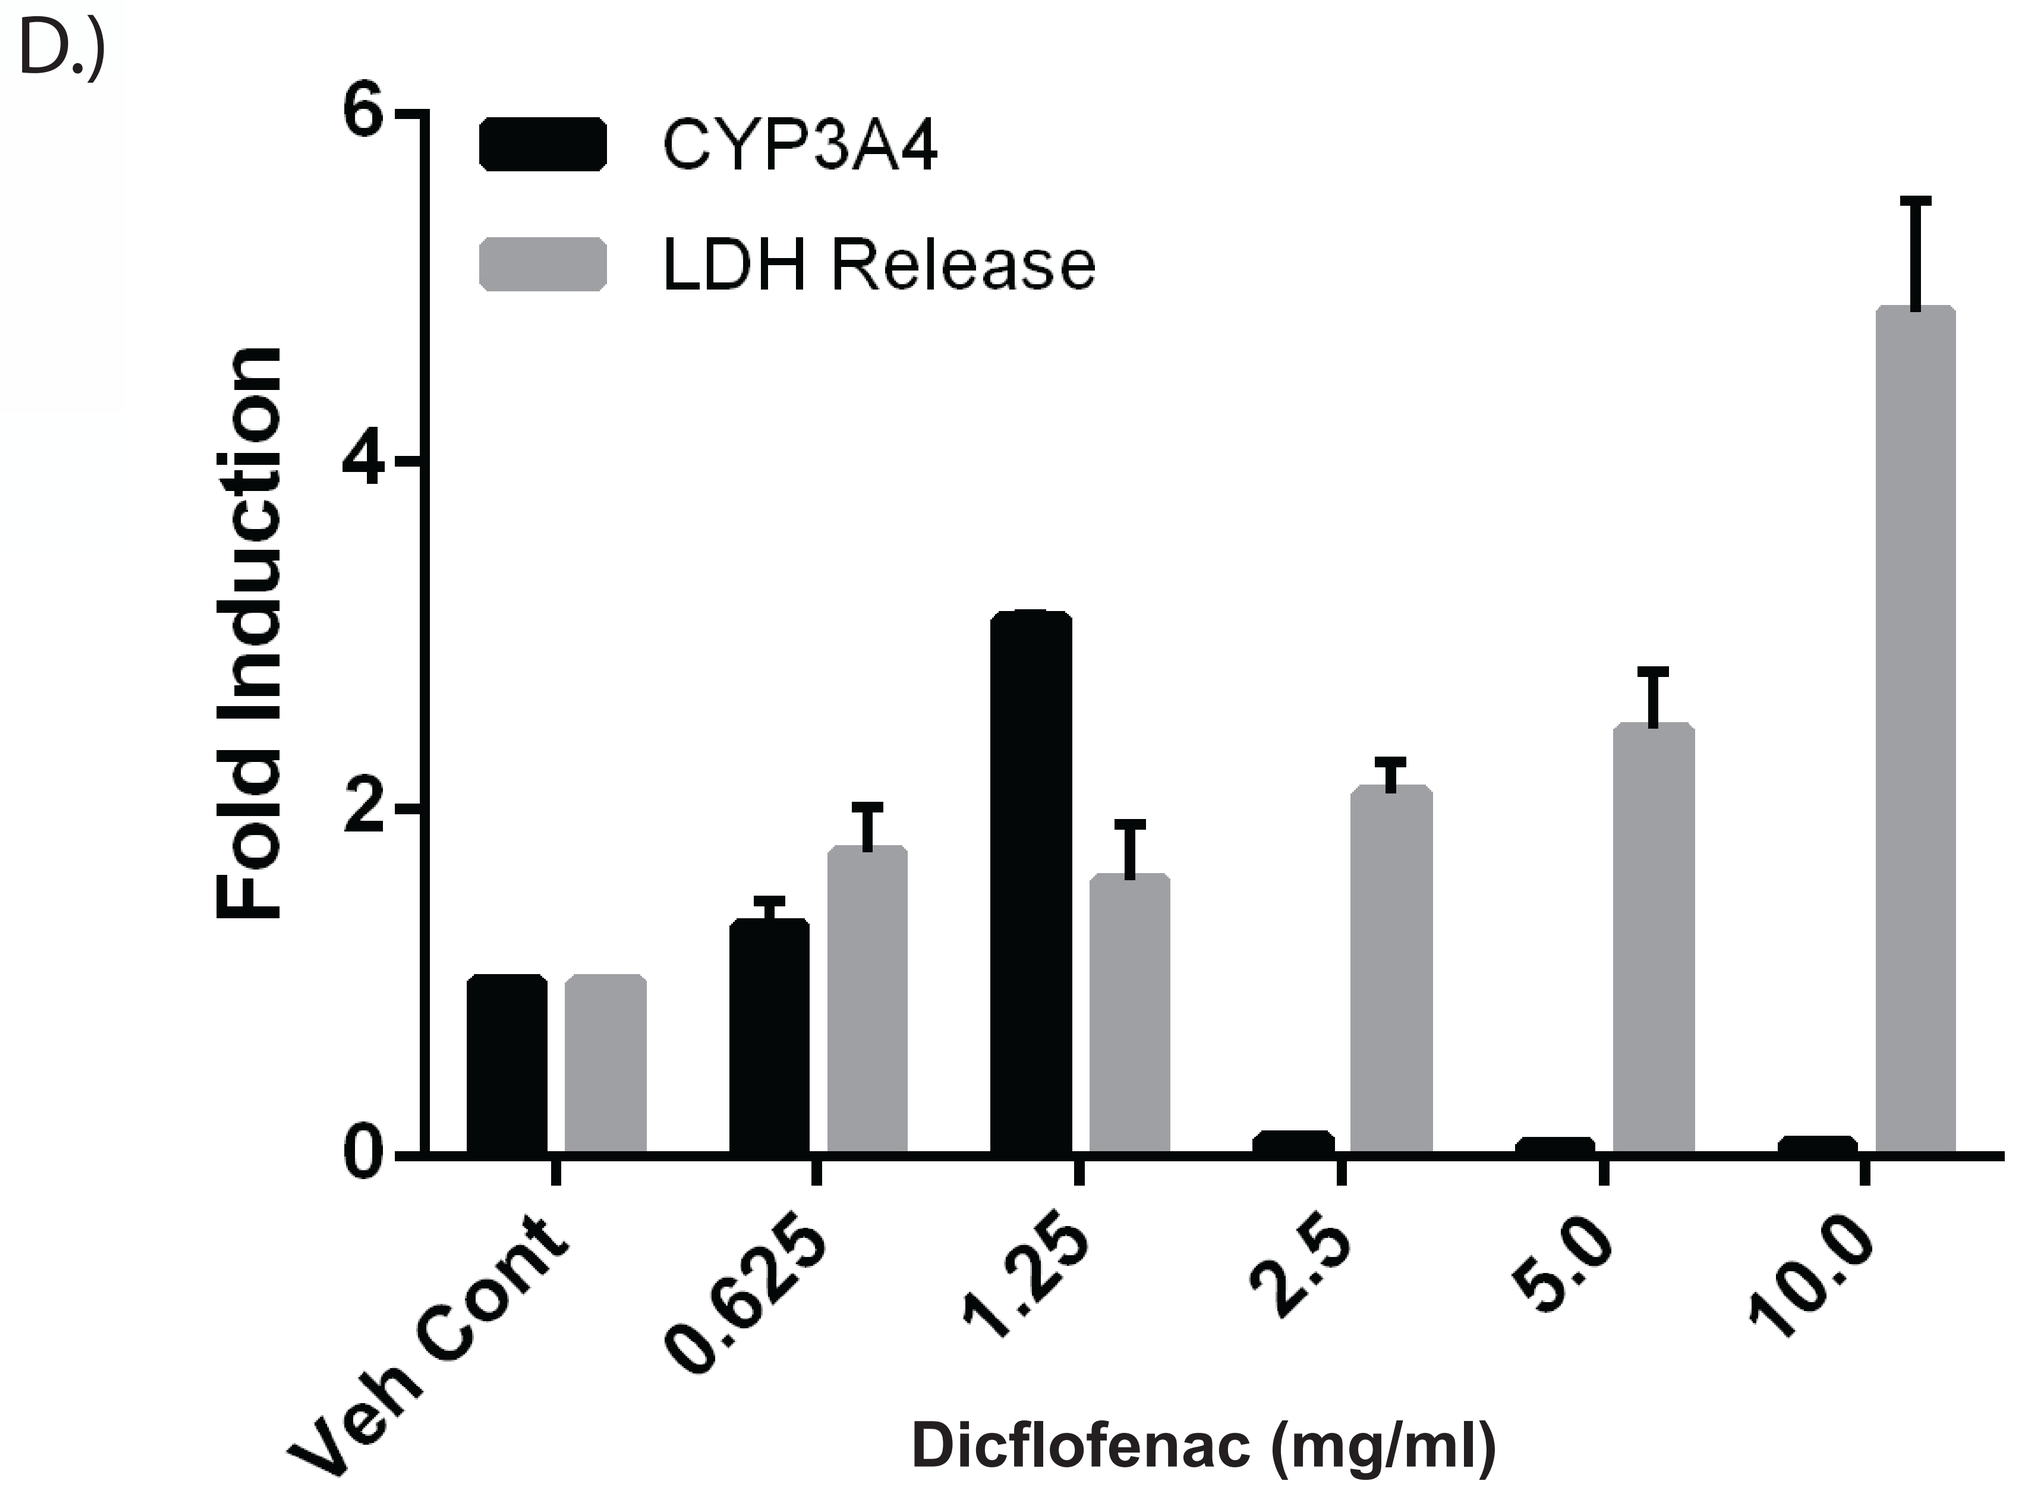
D.)
CYP3A4
LDH Release
Fold Induction
0
2
4
6
Veh Cont
0.625
1.25
2.5
5.0
10.0
Dicflofenac (mg/ml)

Figure 4. 3D bioprinted tissues possess liver-associated features and function for extended time periods. Bioprinted tissues demonstrated tissue-like tight junctions as measured by E-cadherin (A.) and Claudin (B.) stains at day 3. C.) Perfomance of 3D liver tissues over 14 days was validated by measuring albumin production over time. D.) Liver tissues treated with diclofenac after incubating for 14 days in culture, demonstrate the expected induction of CYP3A4 at moderate concentrations, while at high concentrations, damage is apparent by lactate dehydrogenase (LDH) induction and concomitant decrease in CYP3A4.

Results

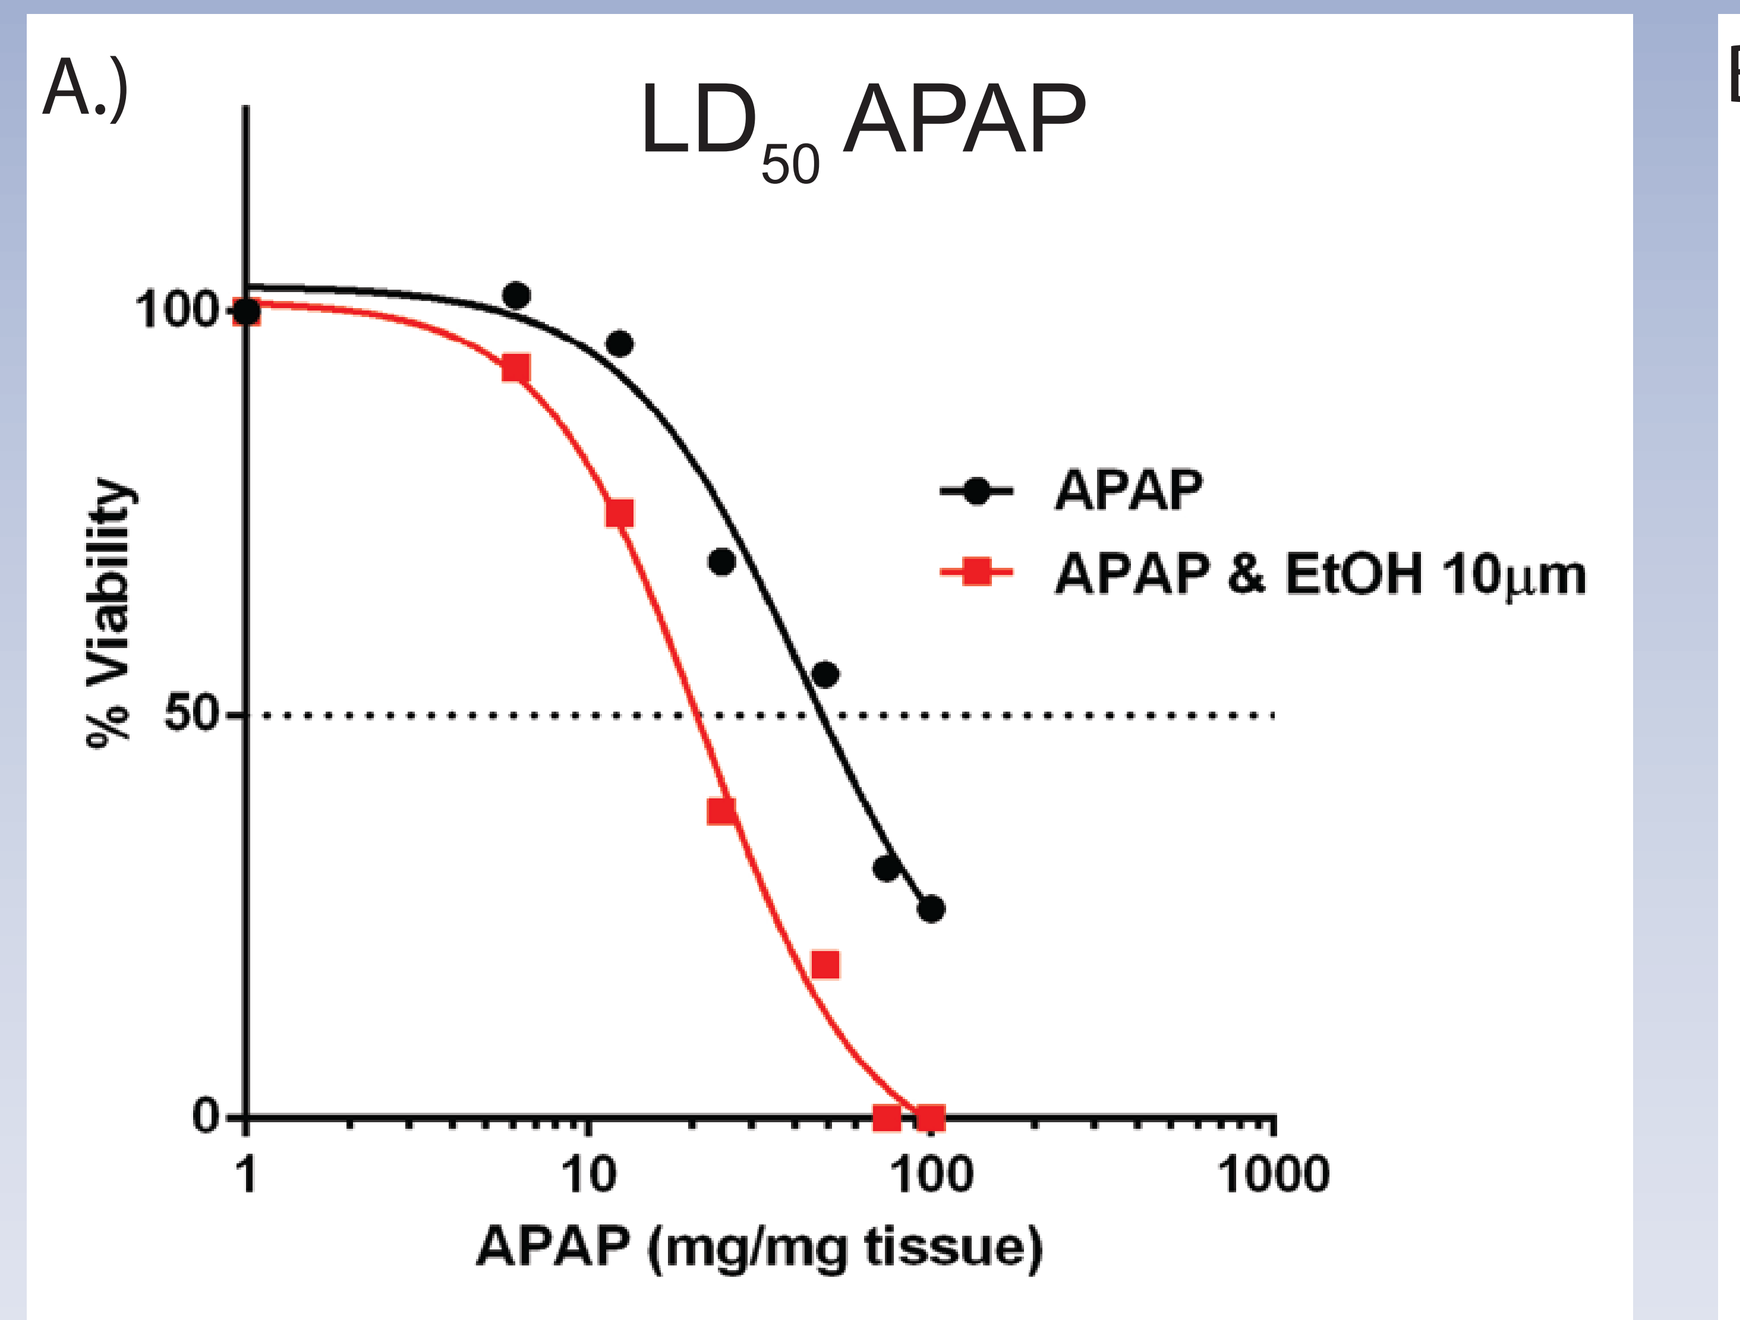
A.)
LD50 APAP
% Viability
0
50
100
APAP
APAP & EtOH 10µm
1
10
100
1000
APAP (mg/mg tissue)

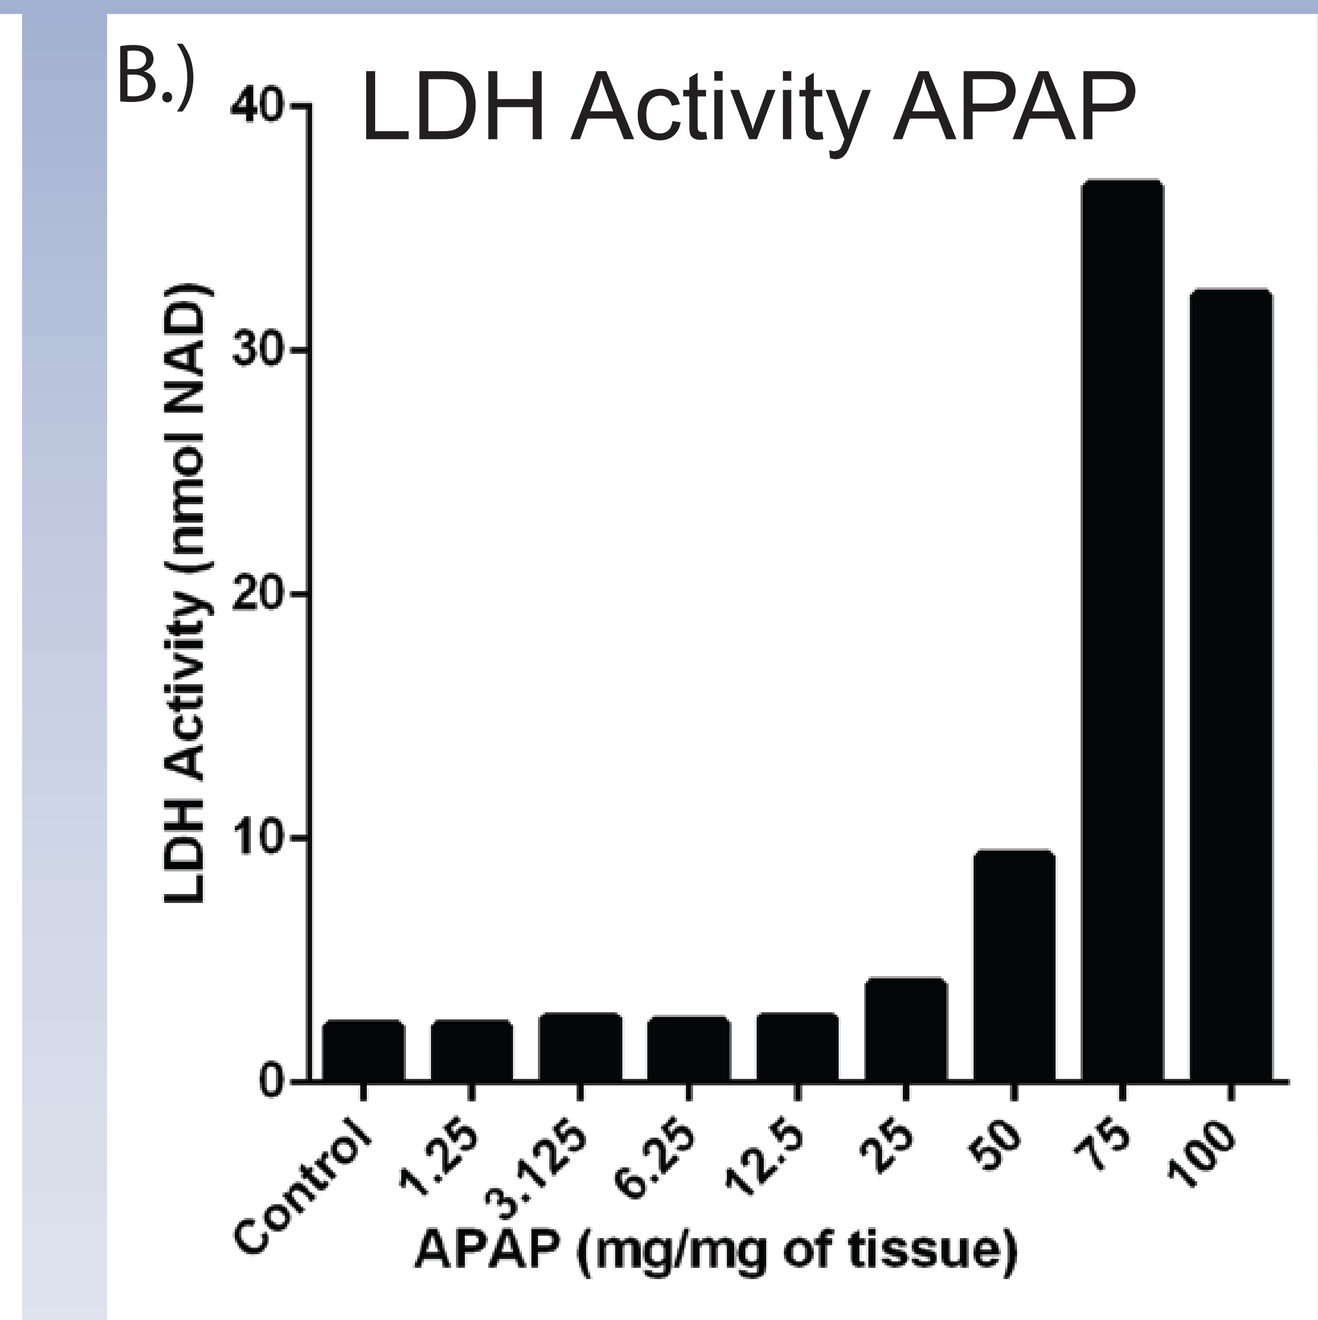
B.)
LDH Activity APAP
LDH Activity (nmol NAD)
0
10
20
30
40
Control
1.25
3.125
6.25
12.5
25
50
75
100
APAP (mg/mg of tissue)

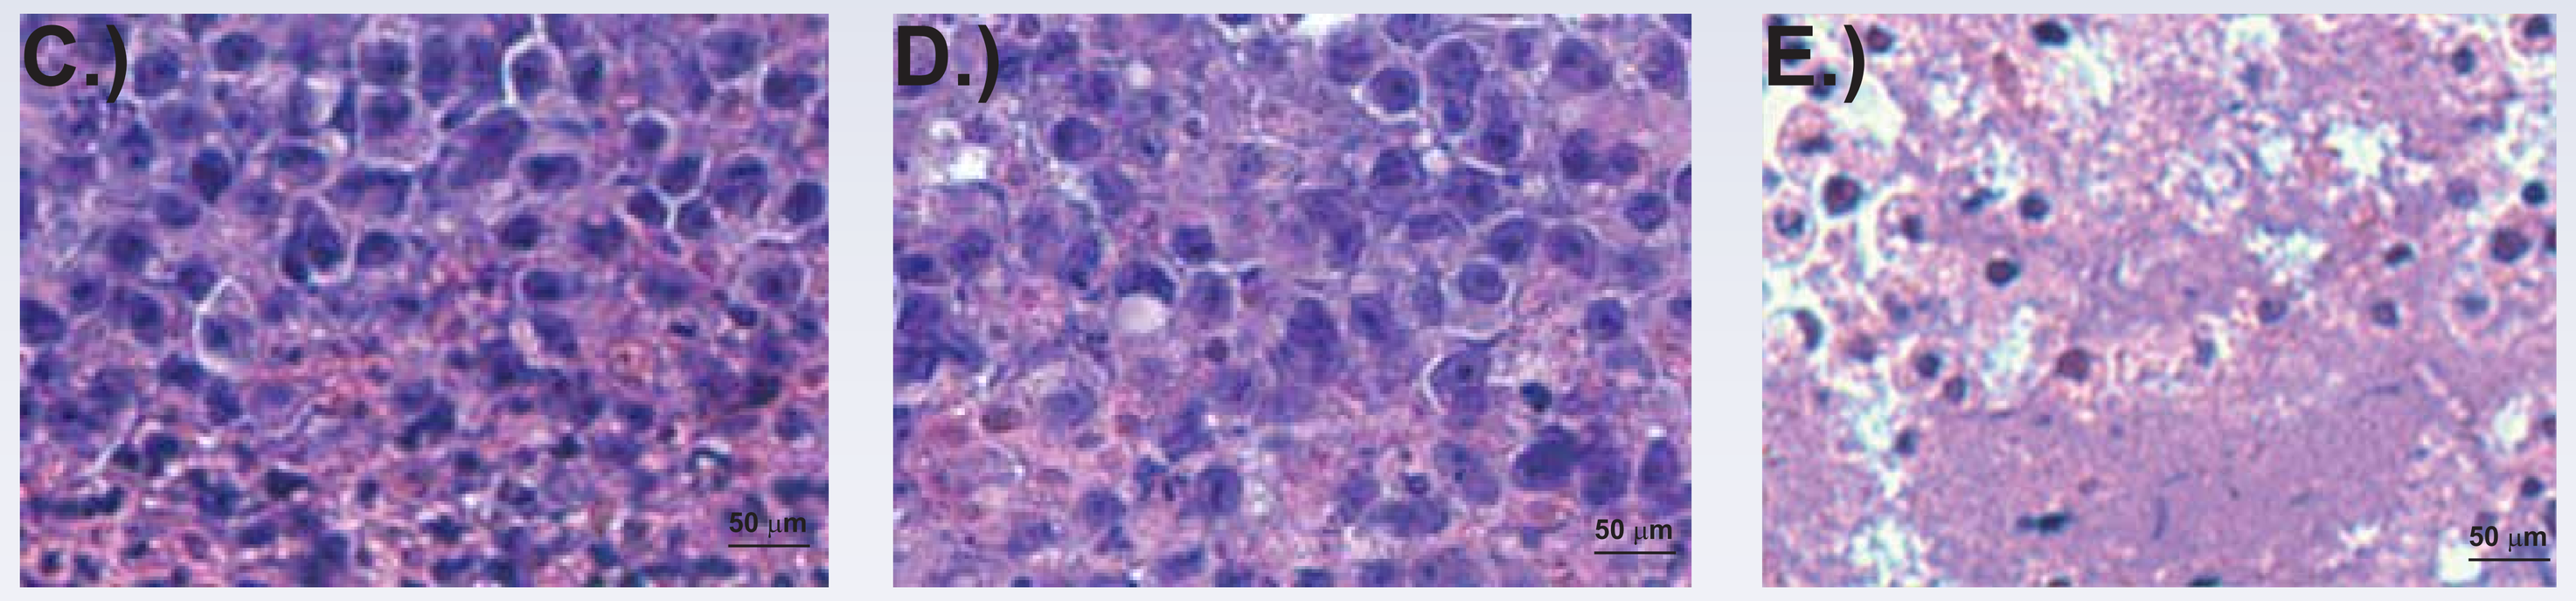
C.)
D.)
E.)
50 µm
50 µm
50 µm

Media Control Vehicle Control APAP (75 mg/mg)

Figure 5. Bioprinted 3D liver tissues can effectively model hepatotoxic insults. A.) Viability of liver tissue is diminished by treatment with acetaminophen (APAP) +/- ethanol (EtOH), as shown by a dose dependent decrease in ATP (Cell Titer Glo; Promega). Addition of EtOH shifts the LD_{50} approximately 2 fold. B.) Increases in LDH indicate tissue damage in the presence of increasing doses of APAP. H&E staining of liver tissues (non-treated controls (C. and D.)) shows pronounced necrosis in those treated with APAP (E.).

Conclusions

Organovo's unique bioprinted 3D liver tissues exhibit several key attributes including:

- Cellular composition closely mimicking human liver tissue
- Development of well organized, tissue-like architecture >250 µm in thickness
- Sustained metabolic activities over time
- Demonstrated response to known hepatotoxic agents (diclofenac and APAP)
- Reproducible, multi-well format

These data further support the use of 3D bioprinted liver tissues as a novel system to interrogate liver biology both biochemically and histologically.

Acknowledgements

The authors would like to thank our colleagues Vina Keophilaone, Jeff Nickel, Olivia Creasey, and Shelby King for excellent histological technical assistance.

Safe Harbor Statement

Any statements contained in this report and presentations that do not describe historical facts may constitute forward-looking statements as that term is defined in the Private Securities Litigation Reform Act of 1995. Any forward-looking statements contained herein are based on current expectations, but are subject to a number of risks and uncertainties. The factors that could cause actual future results to differ materially from current expectations include, but are not limited to, risks and uncertainties relating to the Company's ability to develop, market and sell products based on its technology; the expected benefits and efficacy of the Company's products and technology; the market acceptance for the Company's products, and the risks related to the Company's business, research, product development, regulatory approval, marketing and distribution plans and strategies. These and other factors are identified and described in more detail in the Company's filings with the SEC, including its prospectus supplement filed with the SEC on November 27, 2013, its report on Form 10-Q filed November 8, 2013 and its transition report on Form 10-KT filed with the SEC on May 24, 2013 and our other filings with the Securities and Exchange Commission. You should not place undue reliance on these forward-looking statements, which speak only as of the date of this Current Report. These cautionary statements should be considered with any written or oral forward-looking statements that we may issue in the future. Except as required by applicable law, including the securities laws of the United States, we do not intend to update any of the forward-looking statements to conform these statements to reflect actual results, later events or circumstances or to reflect the occurrence of unanticipated events.